Therapeutic targeting of MARCO with PY265 antibody promotes myeloid cell reprogramming and unleashes anti-tumor immunity

Linnea Haeggblom, Subhadra Dash, Yelena Vayn, Josh Rudolph, Rachael Palmer, Xi Yang, Cesar Juarez, Rachel Greathouse, Carlos Santamaria, Maryam Yousefi, Hanna Ramoth, Ranna Mehta, Erin Mayes, Chris Brassell, Svetlana Gratch, Shilpa Mankikar, Manith Norng, Amy-Jo Casbon, Joshua Pollack, Marc Chamberlain, Leonard Reyno, Matthew Myers, Vladislava Juric, Linda Liang, Kristen Pierce, Kevin P. Baker, and Nadine Jahchan

Pionyr Immunotherapeutics, 2 Tower Place, Suite 800, South San Francisco, CA 94080

PIONYR IMMUNOTHERAPEUTICS

PionyrTX.com

Abstract

Background: The tumor microenvironment (TME) contains immunosuppressive myeloid cells that contribute to checkpoint inhibitor (CPI) resistance. One approach of Pionyr Immunotherapeutics' Myeloid Tuning strategy is to reprogram immunosuppressive myeloid cells to acquire an immunostimulatory anti-tumor phenotype. Macrophage receptor with collagenous structure (MARCO) is an attractive target for myeloid reprogramming, considering its immunomodulatory function, and its expression on tumor-associated macrophages (TAMs) and monocytic myeloid-derived suppressor cells (mMDSCs) in the TME. Pionyr has developed a humanized IgG1k anti-MARCO monoclonal antibody (mAb), PY265, to investigate the potential of MARCO modulation as an anti-cancer immunotherapeutic strategy.

Methods: MARCO expression on TAMs and mMDSCs from multiple solid tumor indications was determined by single-cell RNA sequencing (scRNAseq) and immunohistochemistry (IHC). PY265 levels and activity on human monocyte-derived macrophages (hMDMs) were evaluated by transcriptional profiling, phosphoprotein array, flow cytometry, and measurements of cytokines and chemokines. In vivo efficacy and pharmacodynamic (PD) studies were performed with a surrogate anti-mouse MARCO antibody (PY265m) as single agent or in combination with anti-PD-1 in mice with syngeneic tumors.

Results: MARCO is specifically enriched in a cluster of TAMs and mMDSCs that correlates with immunosuppressive signatures in different types of human solid tumors. IHC studies have shown that MARCO is often expressed in the TME, in metastatic lesions, and in chemo- and CPI-treated tumors. PY265 induces reprogramming in hMDMs in vitro, through induction of rapid phosphorylation events, transcriptional activation of pro-inflammatory pathways, production of cytokines and chemokines, and upregulation of cell surface activation receptors. PY265m has significant anti-tumor activity in syngeneic mouse models, as a single agent in CPI-sensitive models, and in combination with anti-PD-1 in CPI-resistant models. PD studies suggest that PY265m induces immune activation by reprogramming pro-tumorigenic, M2-like TAMs and mMDSCs to proinflammatory anti-tumor M1-like macrophages and monocytes, leading to increased infiltration of the tumor, spleen, and tumor-draining lymph nodes by effector cells. In a non-human primate toxicokinetic study, PY265 was generally well tolerated at all dose levels tested.

Conclusions: Our studies demonstrate that targeting MARCO reprograms myeloid cells and remodels the TME to unleash anti-tumor immunity and convert CPI-resistant tumors into treatment-responsive tumors. Collectively, these preclinical data support PY265 immunotherapy, alone or in combination with a CPI, in patients with cancer resistant or refractory to CPI therapies, to potentially improve rates of overall response and the durability of response. First-in-human testing of PY265 will be initiated in 2023.

Targeting the MARCO Receptor with PY265

MARCO: **M**acrophage **R**eceptor with **Co**llagenous Structure

Function: scavenger and immunomodulatory receptor
- Defense against microbial pathogens via binding to ligands including foreign polyanionic ligands, bacteria, and endogenous lipoproteins
- Role in macrophage phagocytosis, adhesion, migration, and TLR-induced activation

Genetics: MARCO-knockout mice are viable and fertile, but have increased susceptibility to bacterial infection and inhaled particles, impaired macrophage phagocytosis, and abnormal spleen marginal zone morphology

Relevance for Immuno-Oncology
- MARCO is enriched on immunosuppressive TAMs and mMDSCs, and is upregulated in response to IL-10 and TGF-β
- Targeting MARCO reprograms TAMs and monocytes from an immunosuppressive to a proinflammatory phenotype and restores the cytotoxic properties of NK and T cells

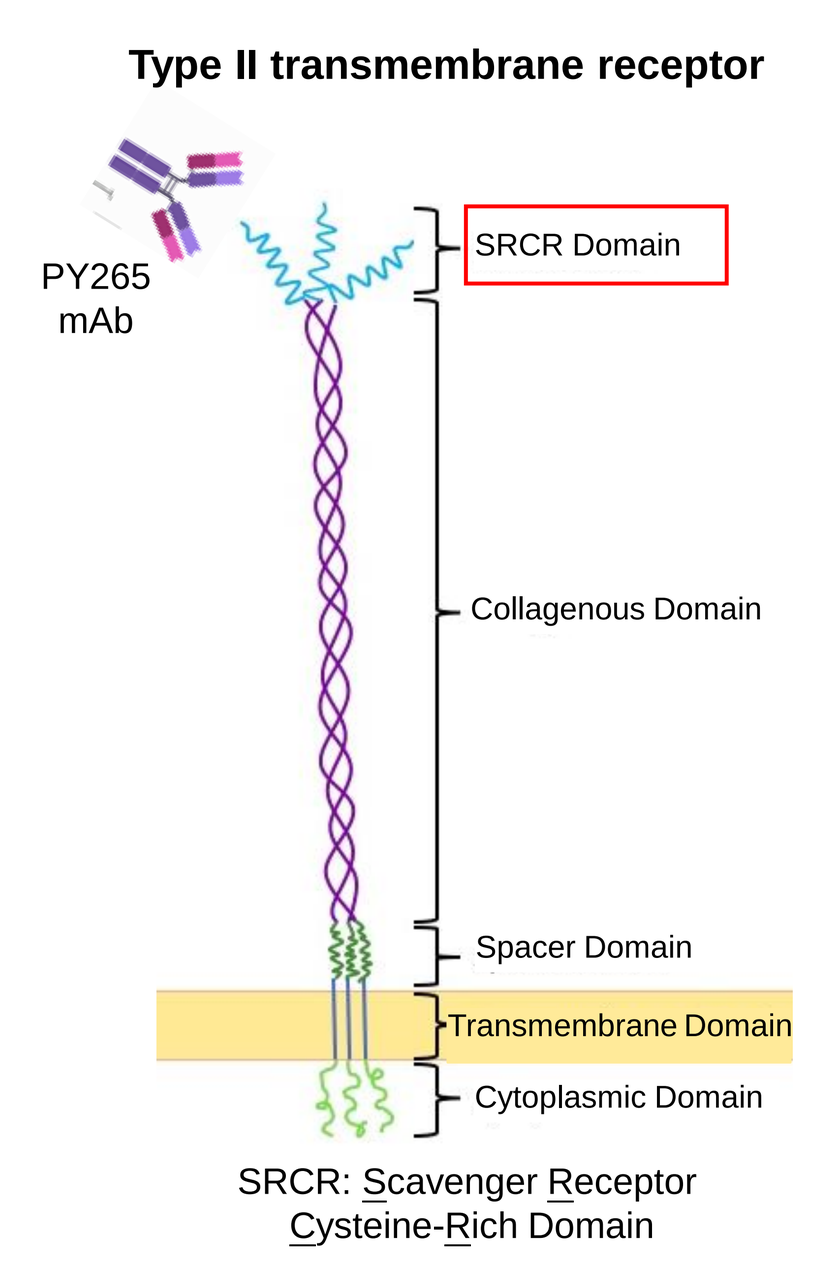
Type II transmembrane receptor
PY265 mAb
SRCR Domain
Collagenous Domain
Spacer Domain
Transmembrane Domain
Cytoplasmic Domain
SRCR: Scavenger Receptor Cysteine-Rich Domain

PY265: a humanized IgG1 mAb that binds the SRCR domain of human- and cyno-MARCO
PY265m: a mouse surrogate IgG2a mAb that binds the SRCR domain of murine MARCO

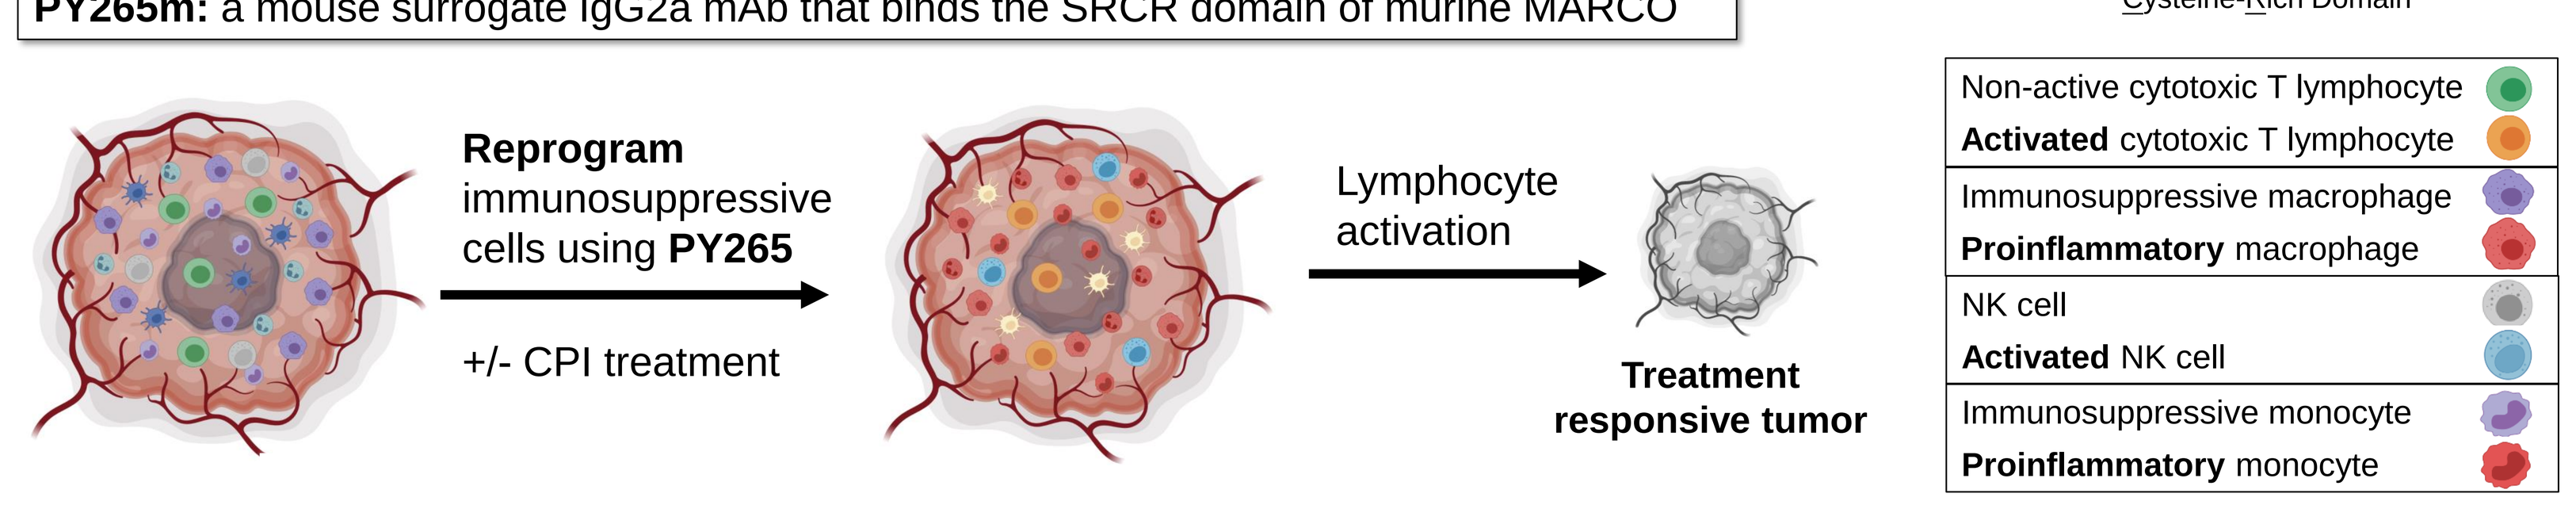
Reprogram immunosuppressive cells using PY265
+/- CPI treatment
Lymphocyte activation
Treatment responsive tumor
Non-active cytotoxic T lymphocyte
Activated cytotoxic T lymphocyte
Immunosuppressive macrophage
Proinflammatory macrophage
NK cell
Activated NK cell
Immunosuppressive monocyte
Proinflammatory monocyte

Figure 1. Pionyr's Myeloid Tuning approach involves modulating inhibitory myeloid populations in the TME with high precision and selectivity. PY265 (anti-MARCO) "re-tunes" the TME by reprogramming the immunosuppressive myeloid cells to acquire a proinflammatory phenotype and generate effective anti-tumor immunity.

MARCO-Expressing Monocytes and Macrophages in Human Tumors Have an Immunosuppressive Signature and Correlates with Poor Survival

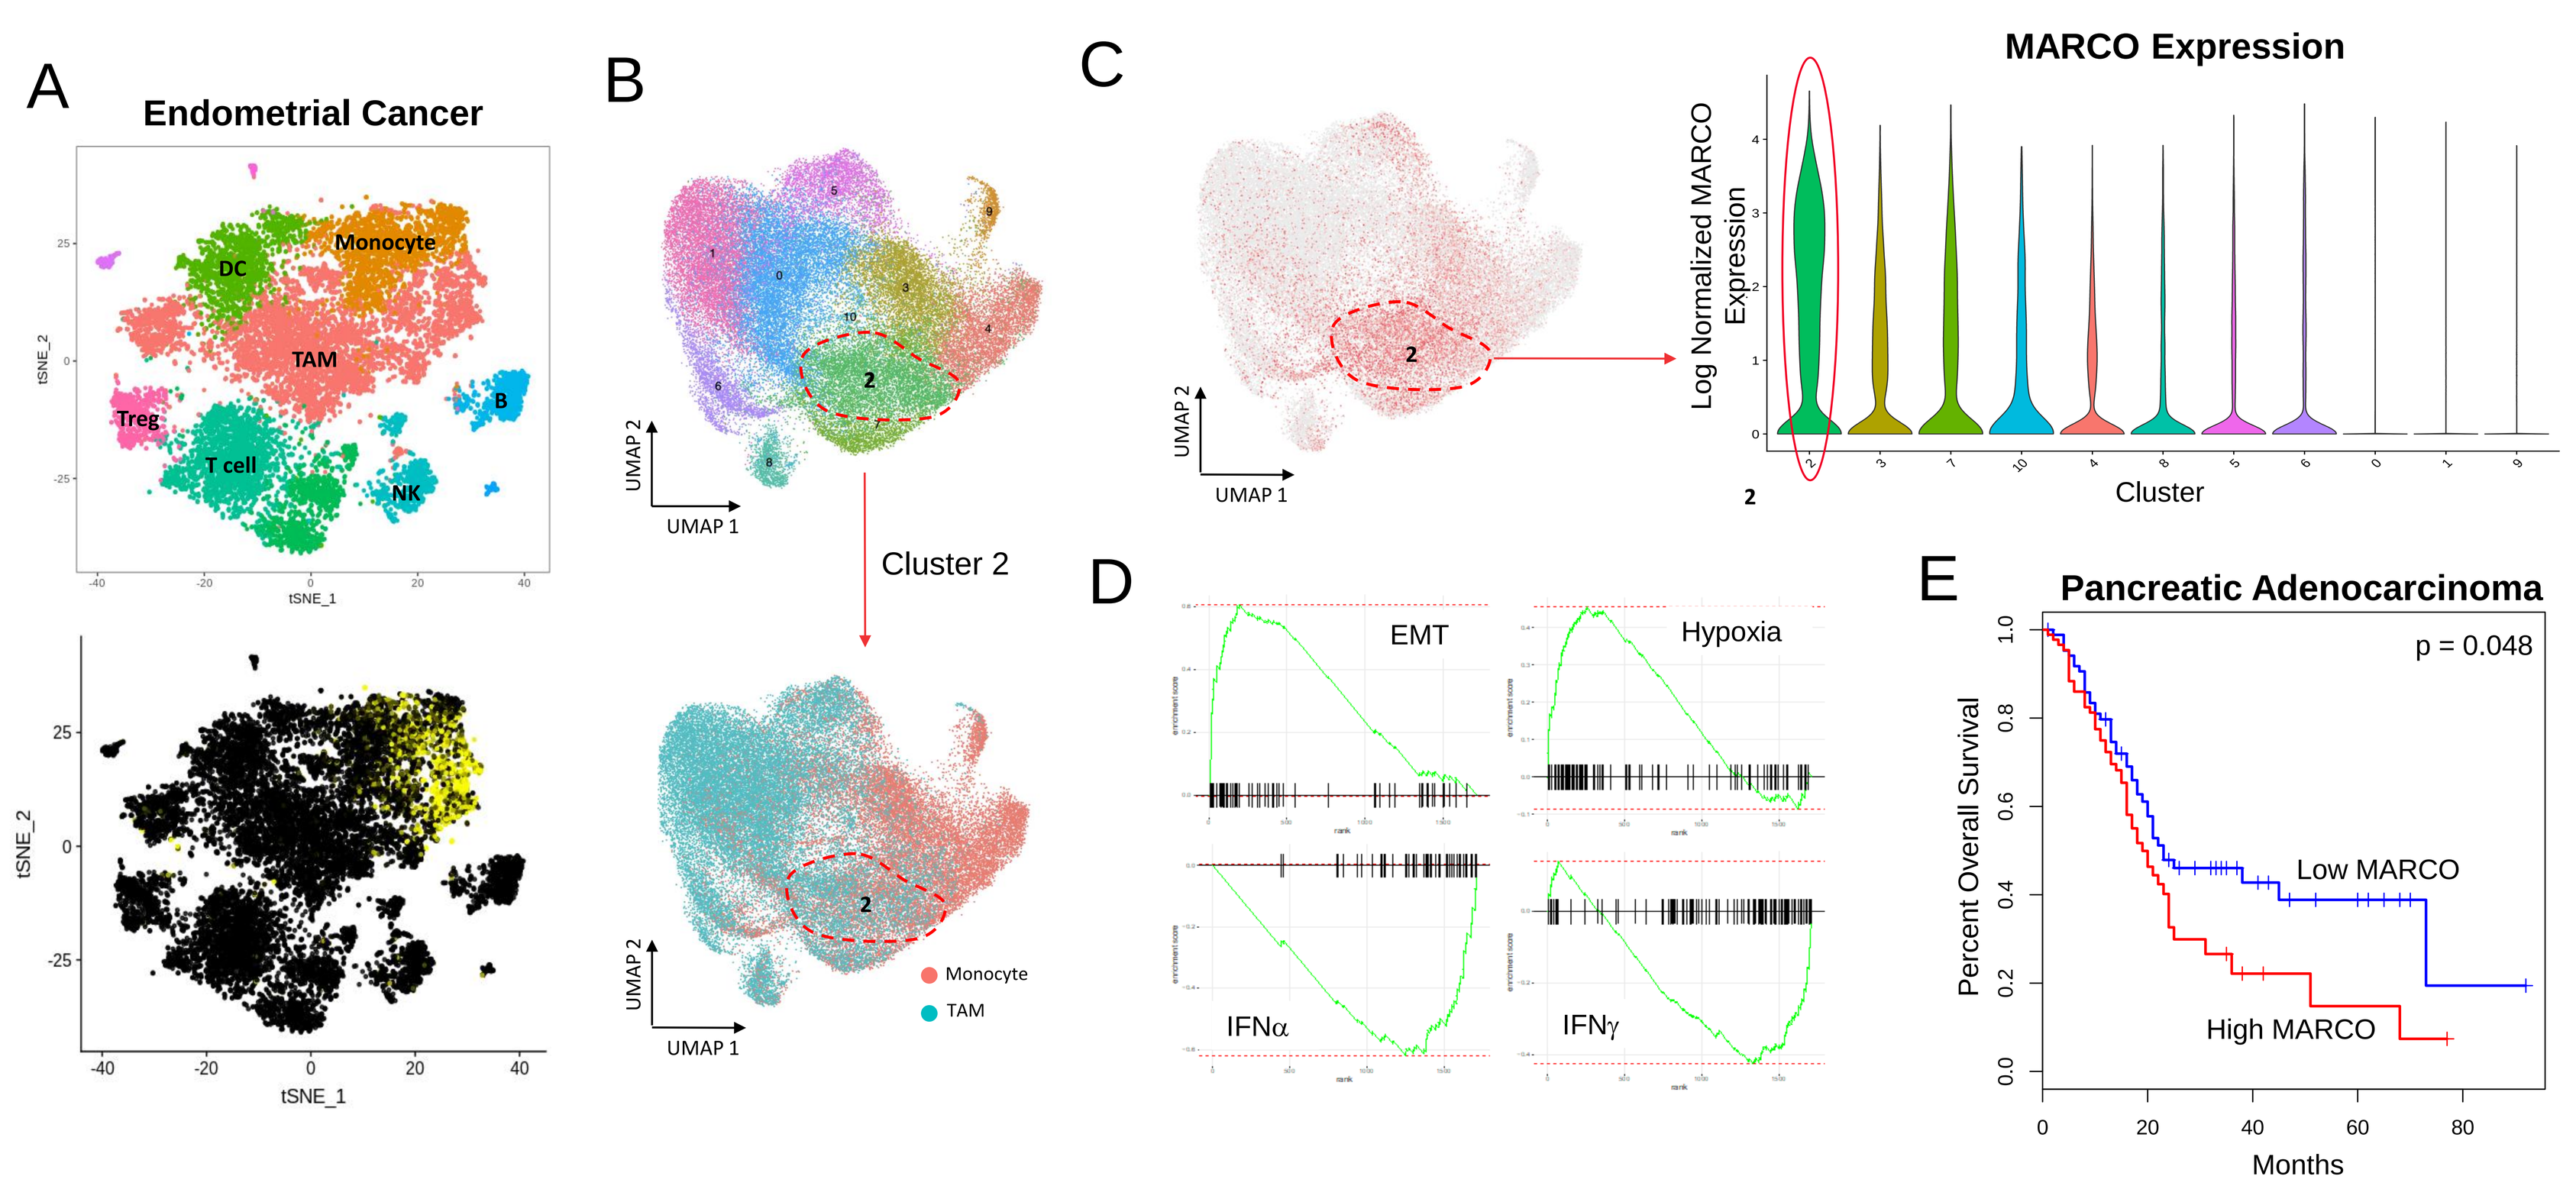
A
Endometrial Cancer
DC
Monocyte
TAM
Treg
T cell
NK
B
tSNE_1
tSNE_2
B
UMAP 1
UMAP 2
Cluster 2
Monocyte
TAM
C
MARCO Expression
Log Normalized MARCO Expression
Cluster
D
EMT
Hypoxia
IFNα
IFNγ
E
Pancreatic Adenocarcinoma
p = 0.048
Percent Overall Survival
Low MARCO
High MARCO
Months

Figure 2. (**A**) scRNAseq of CD45+ cells from a human endometrial tumor. tSNE plots depict distinct leukocyte subsets (top) and *MARCO* expression in yellow (bottom). **(B)** Aggregated TAMs and monocytes were identified by scRNAseq analyses (Pionyr Immunotherapeutics) of immune cells from 13 samples of different human primary tumors (top). Louvain clustering yielded 11 clusters, where cluster 2 contains intermediate macrophages and monocytes (bottom). **(C)** Cluster 2 has significantly enriched expression of *MARCO* compared with the other clusters **(D)** and correlates with immunosuppressive gene expression signatures (e.g., hypoxia, EMT). Cluster 2 correlates inversely with proinflammatory signatures (e.g., interferon-based pathways), as determined by using Gene Set Enrichment Analysis of hallmark pathways (*MARCO*-rich cluster 2 vs all other macrophages and monocytes). **(E)** Differences in overall survival of patients with pancreatic adenocarcinomas that have above-median (high, red) vs below-median (low, blue) expression of *MARCO* mRNA. Pre-normalized expression values for *MARCO* were determined from the GEPIA, which combines data from TCGA.

MARCO is Expressed in Aggressive Solid Tumors

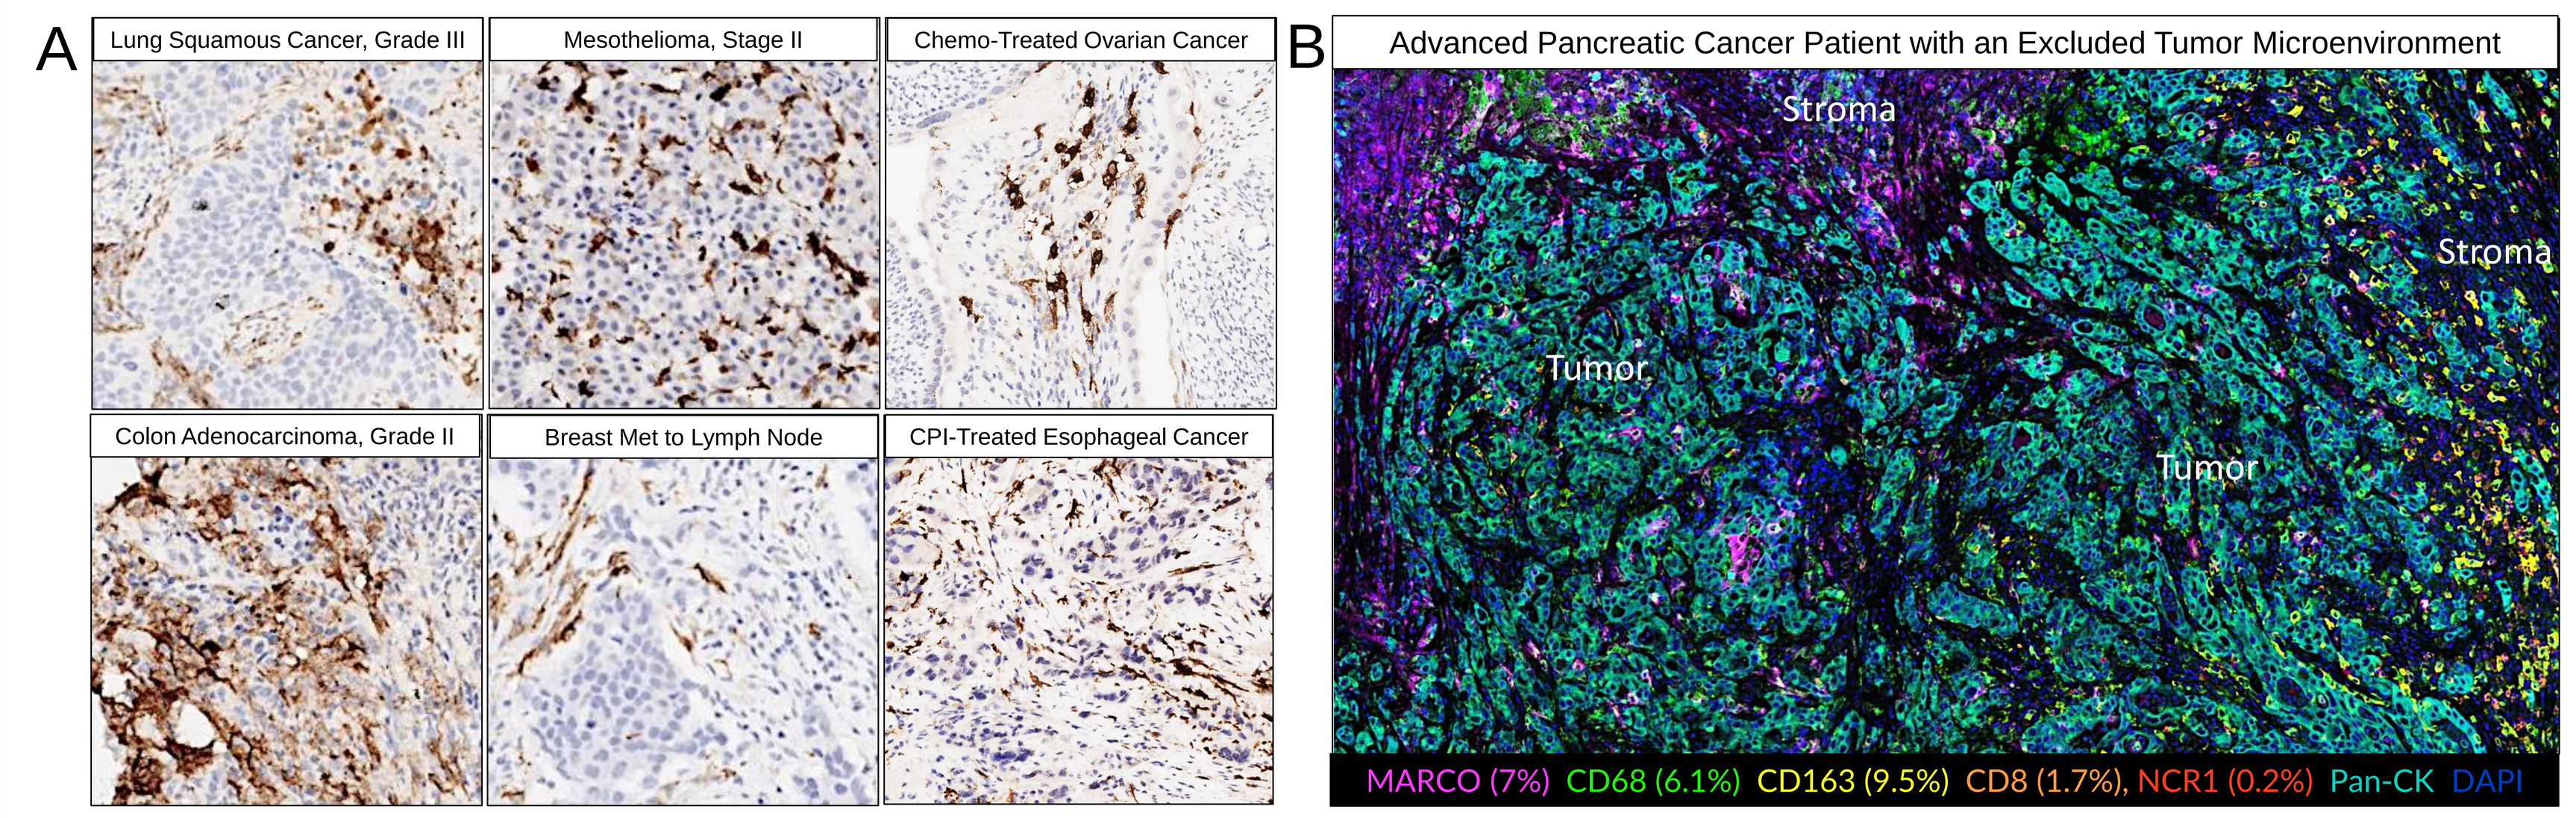
A
Lung Squamous Cancer, Grade III
Mesothelioma, Stage II
Chemo-Treated Ovarian Cancer
Colon Adenocarcinoma, Grade II
Breast Met to Lymph Node
CPI-Treated Esophageal Cancer
B
Advanced Pancreatic Cancer Patient with an Excluded Tumor Microenvironment
Stroma
Stroma
Tumor
Tumor
MARCO (7%) CD68 (6.1%) CD163 (9.5%) CD8 (1.7%), NCR1 (0.2%) Pan-CK DAPI

Figure 3. (A) Representative images of tumor tissues of lung (grade III), mesothelium (stage II), colon (grade II), breast metastasized to lymph node, chemo-treated ovarian, and CPI-treated esophagus, stained with an IHC compatible anti-human MARCO antibody. The brown color indicates myeloid cells expressing MARCO in the tumor stroma. **(B)** Representative image of an advanced pancreatic cancer tissue with an excluded TME (defined by high expression of immune cells in the stroma with little infiltration into tumor nests). MARCO+ myeloid cells (pink), CD68+ (green) and CD163+ (yellow) macrophages, CD8+ cytotoxic T cells (orange), NCR1+ NK cells (red), Pan-CK+ tumor cells (cyan) and DAPI+ nuclei (blue) were stained using multiplex immunofluorescent OPAL IHC technology. HALO image analysis was used to quantify the percentage positive cells per marker from the entire tissue.

PY265 Binds With Specificity and High Affinity to Suppressive hMDMs

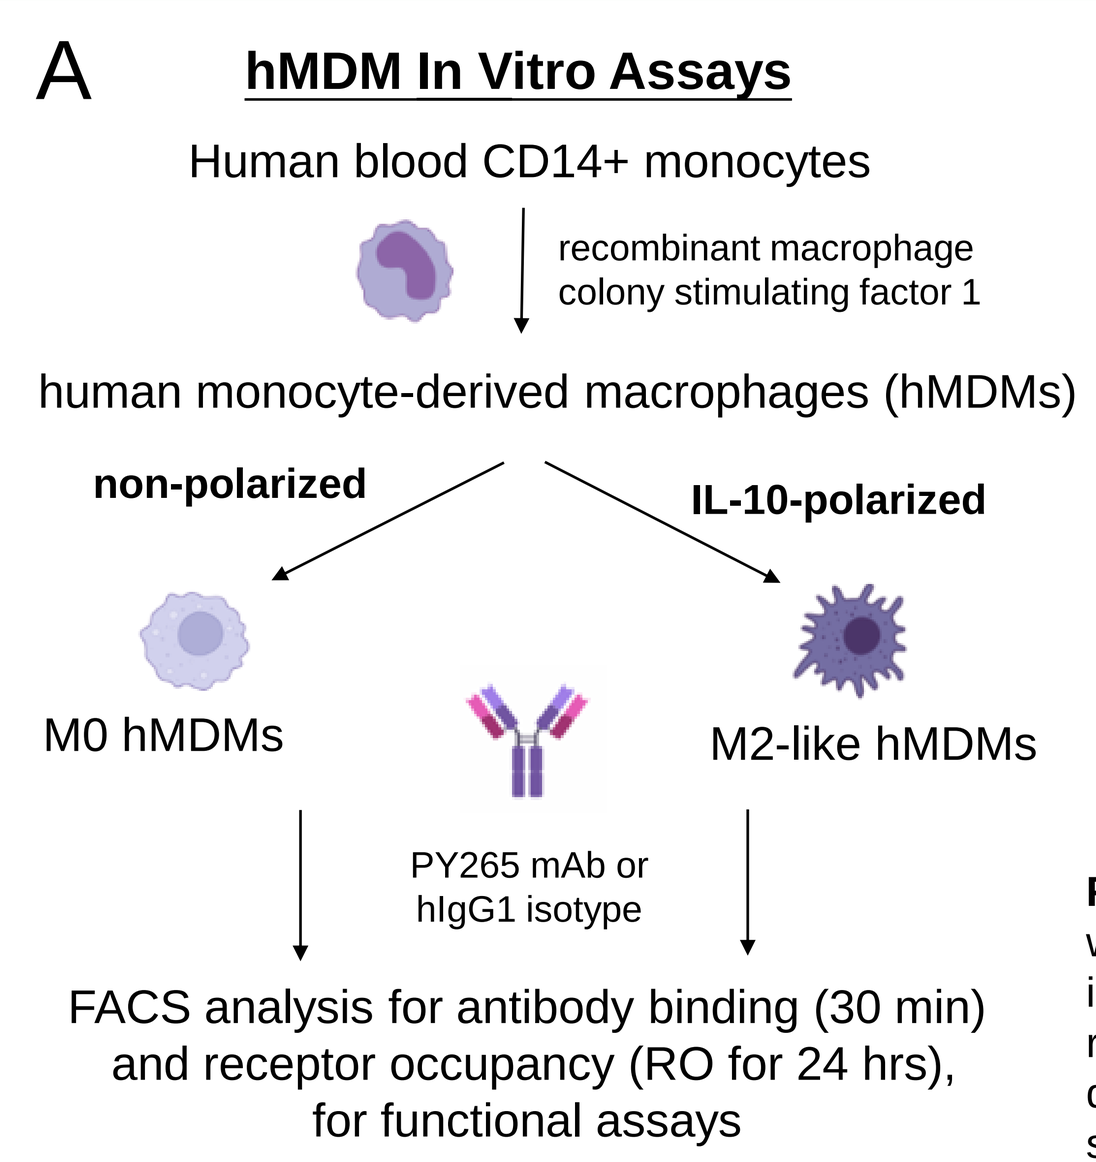
A
hMDM In Vitro Assays
Human blood CD14+ monocytes
recombinant macrophage colony stimulating factor 1
human monocyte-derived macrophages (hMDMs)
non-polarized
IL-10-polarized
M0 hMDMs
M2-like hMDMs
PY265 mAb or hIgG1 isotype
FACS analysis for antibody binding (30 min) and receptor occupancy (RO for 24 hrs), for functional assays

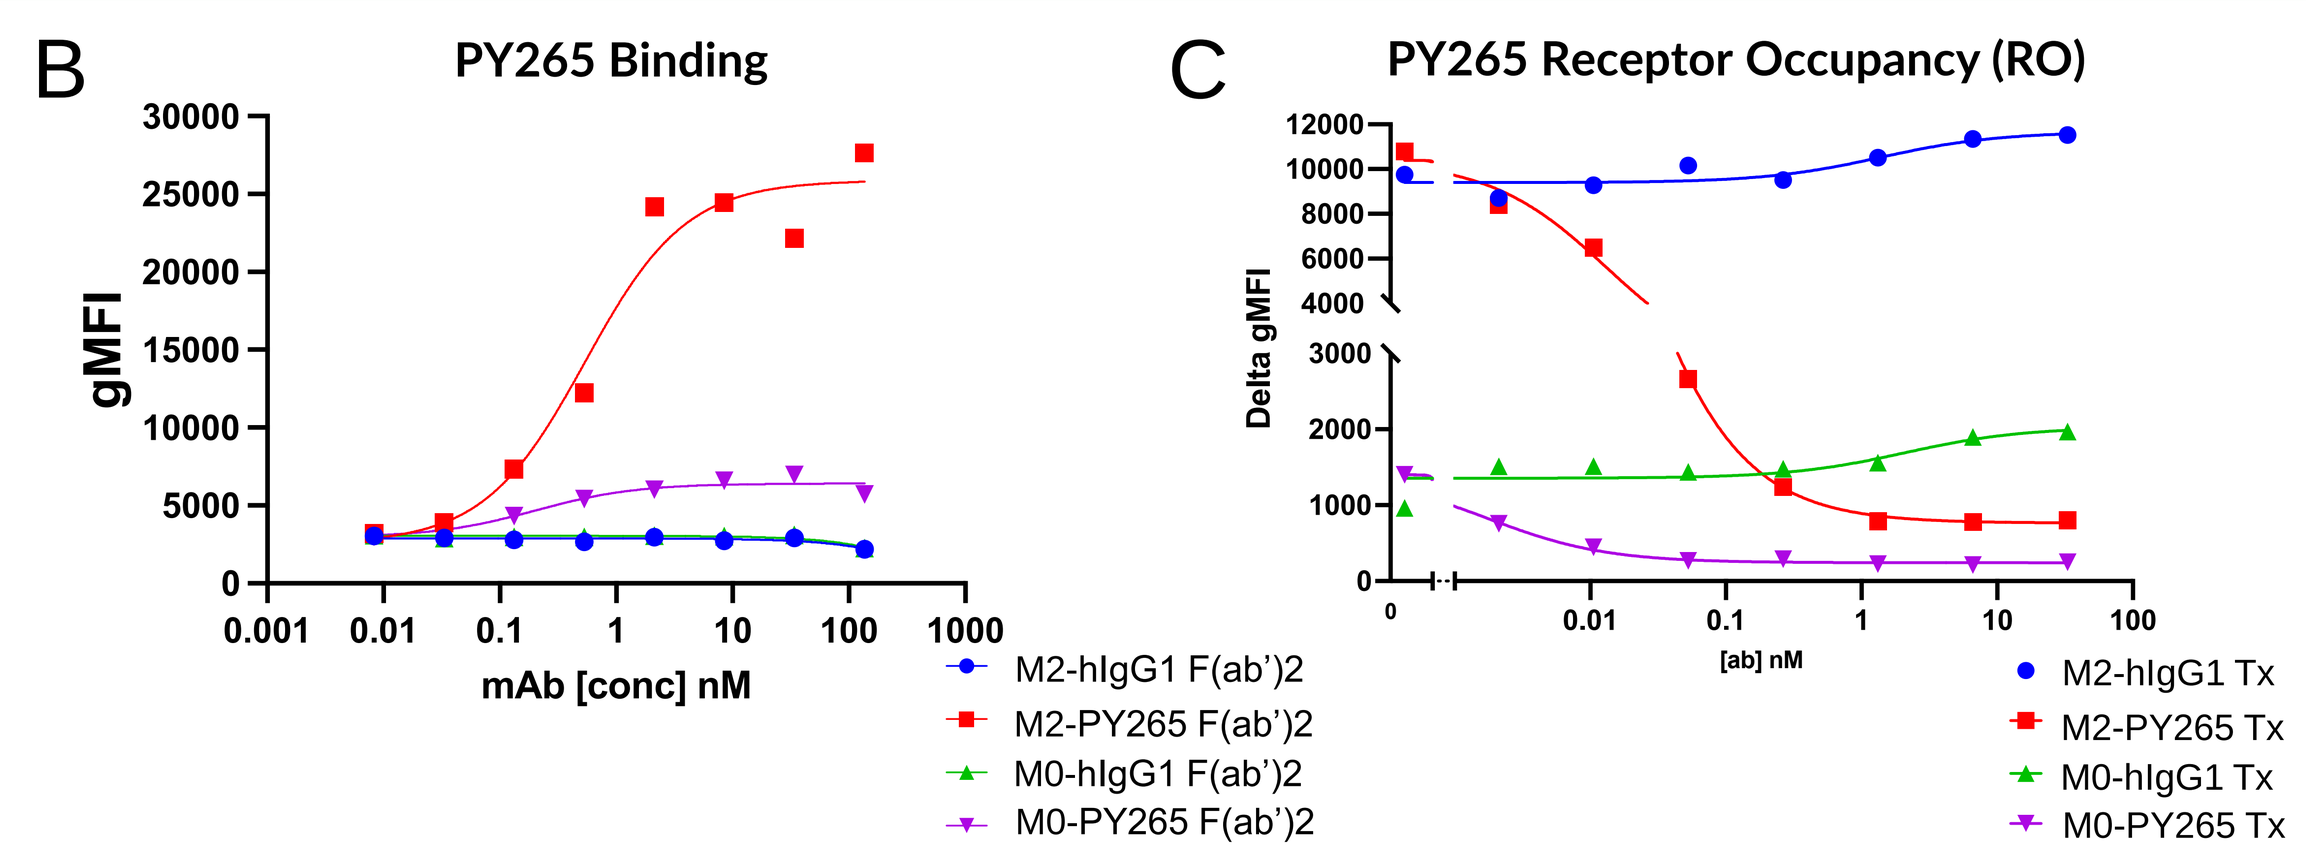
B
PY265 Binding
gMFI
mAb [conc] nM
M2-hIgG1 F(ab')2
M2-PY265 F(ab')2
M0-hIgG1 F(ab')2
M0-PY265 F(ab')2
C
PY265 Receptor Occupancy (RO)
Delta gMFI
[ab] nM
M2-hIgG1 Tx
M2-PY265 Tx
M0-hIgG1 Tx
M0-PY265 Tx

Figure 4. (A) hMDM in vitro assays. **(B)** Non-polarized (M0) or IL10–polarized (M2) hMDMs from 1 representative donor were incubated with increasing doses of PY265 F(ab')2 or hIgG1 F(ab')2. The Y axis shows the geometric mean fluorescent intensity (gMFI) of bound PY265 F(ab')2 and hIgG1 F(ab')2. **(C)** Levels of free MARCO on M2 and M0 macrophages from 1 representative hMDM donor as a function of PY265 and hIgG1 isotype treatment (Tx). The level of free MARCO was determined by staining with a PY265-competing mAb. The y-axis represents delta gMFI of bound PY265 mAb after subtraction of the fluorescence detected with the isotype-matched antibody control.

PY265 Induces Proinflammatory Activation In Vitro

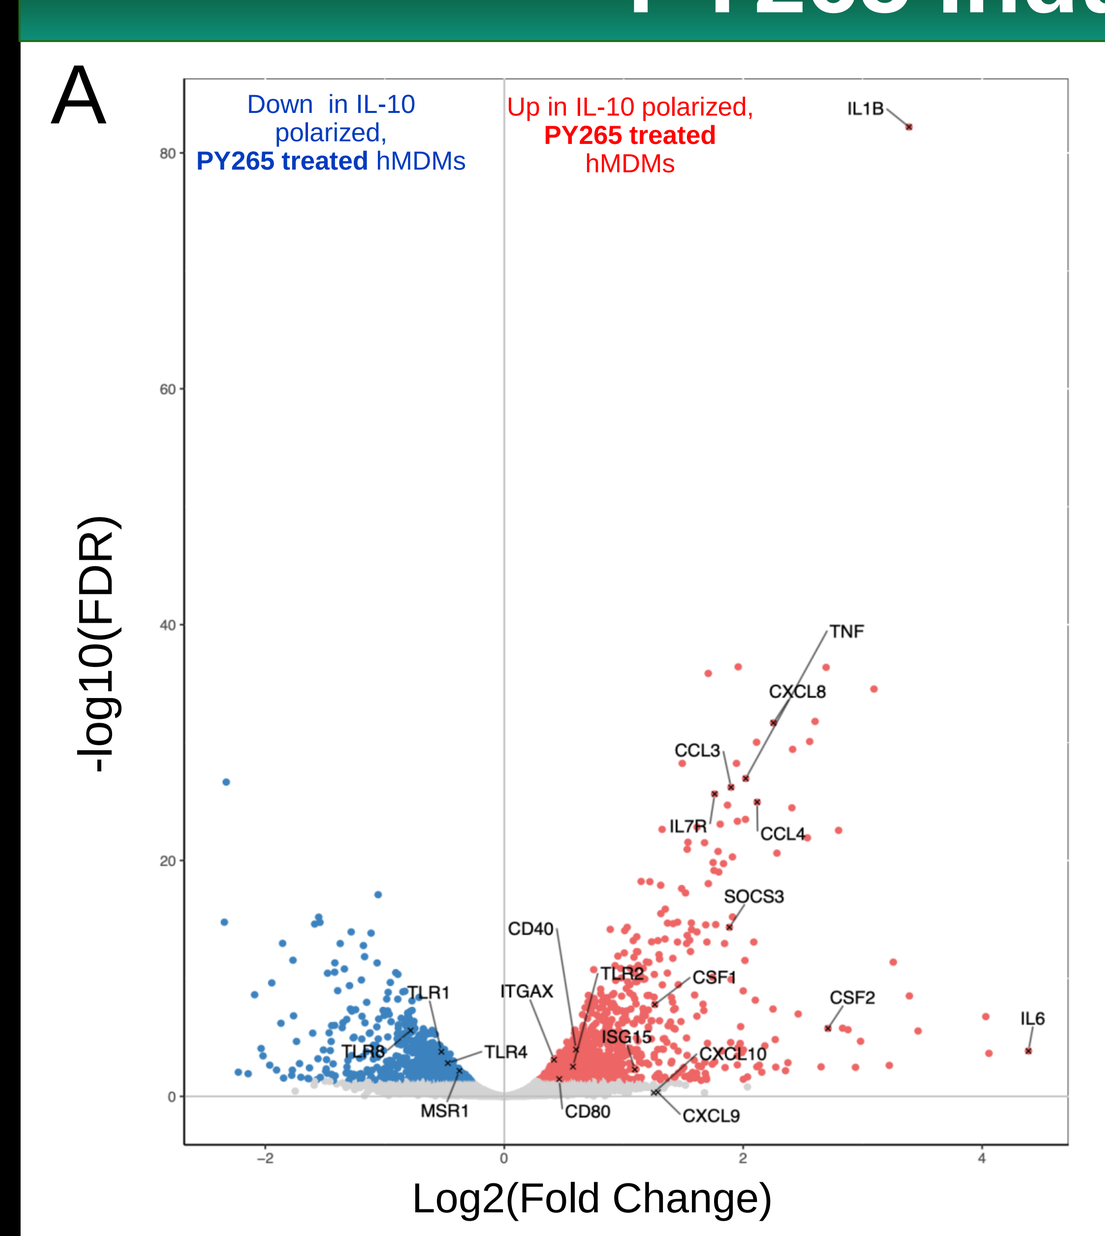
A
Down in IL-10 polarized, PY265 treated hMDMs
Up in IL-10 polarized, PY265 treated hMDMs
-log10(FDR)
Log2(Fold Change)

Significant hallmark pathways induced by PY265 in IL10-polarized hMDMs:
- TNF signaling via NF-κB
- Inflammatory response
- IL-2 STAT5 signaling
- Interferon (IFN)γ response
- IFNα response

Proinflammatory genes induced by PY265:

Cytokines and chemokines:
- IL-1a, CXCL8, IL-6, IL-1b, CCL20, CCL24, CCL3
- Cell surface activation markers: PD-L1, CD11c, CD80, CD40

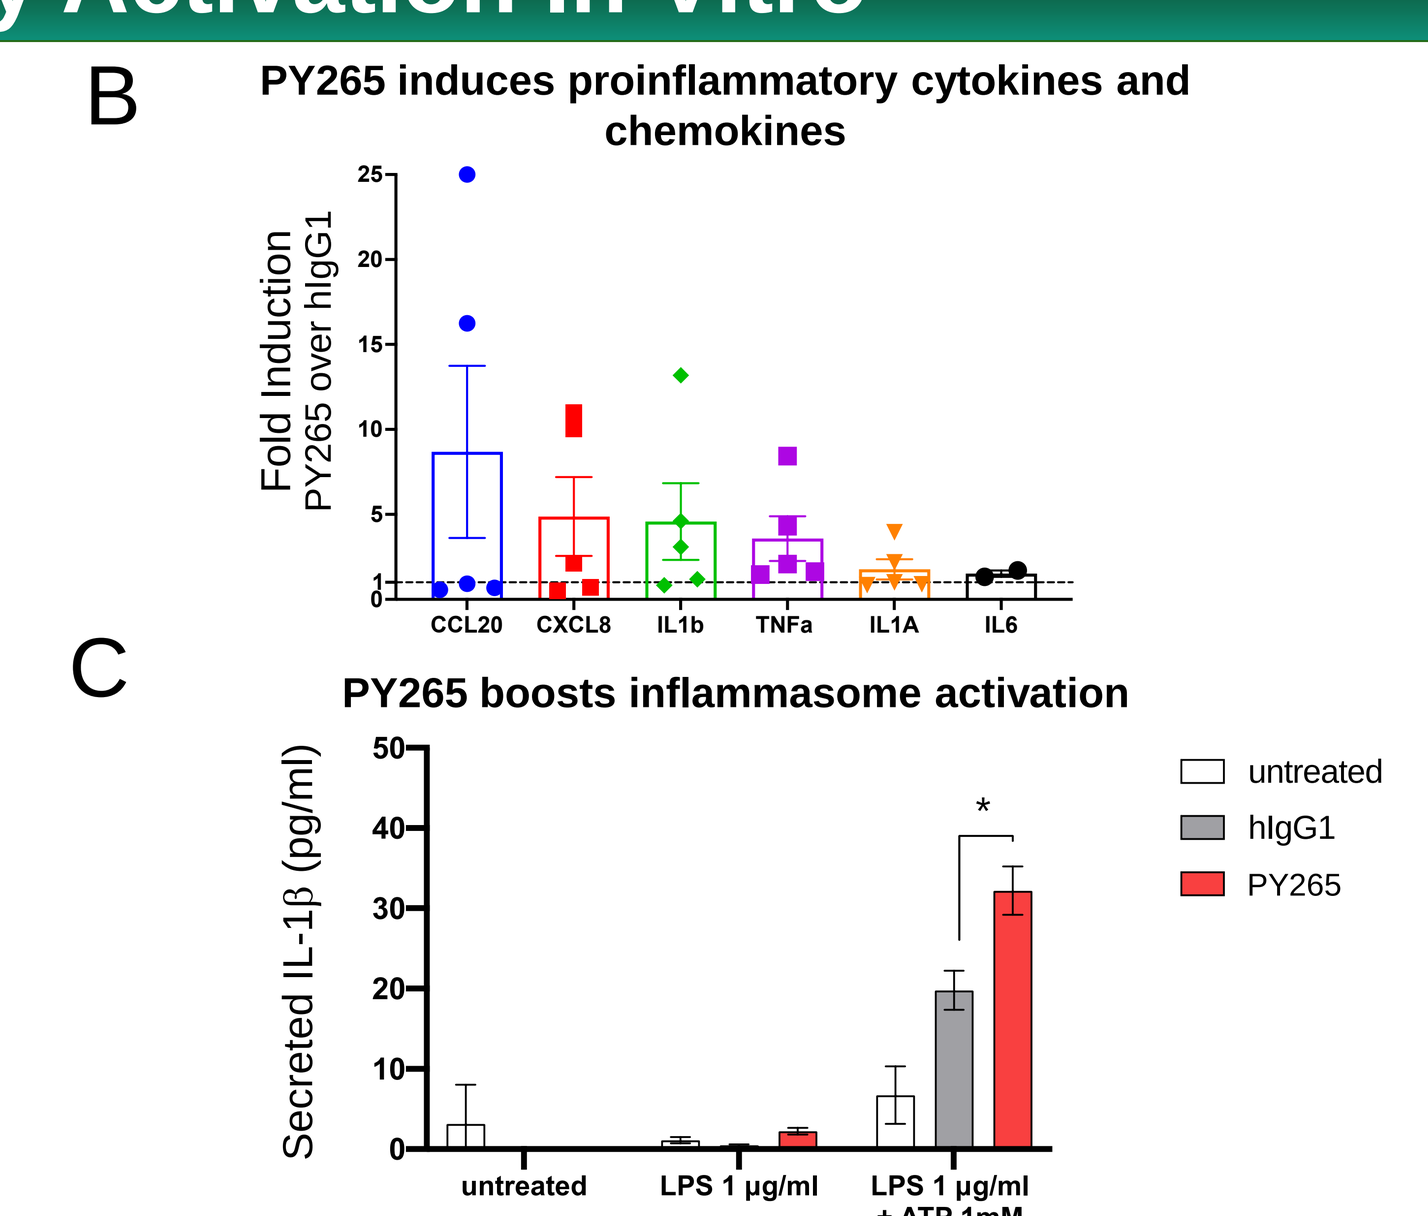
B
PY265 induces proinflammatory cytokines and chemokines
Fold Induction PY265 over hIgG1
CCL20
CXCL8
IL1b
TNFa
IL1A
IL6
C
PY265 boosts inflammasome activation
Secreted IL-1β (pg/ml)
untreated
hIgG1
PY265
untreated
LPS 1 μg/ml
LPS 1 μg/ml + ATP 1mM

Figure 5. (A) RNA sequencing was performed on IL10-polarized hMDMs treated for 4 hrs with PY265 and hIgG1 isotype mAbs. PY265-induced differentially expressed genes and downstream GSEA pathway analyses yielded a profound increase in proinflammatory chemokines/cytokines, activation markers, and a variety of significantly upregulated pathways associated with macrophage activation/reprogramming and inflammatory responses. **(B)** qPCR mRNA analysis of key proinflammatory genes induced after 4 hrs of treatment (1 μg/ml) is plotted as fold induction over the hIgG1 isotype. Data are presented as mean values from 5 donors ± (SEM). **(C)** IL10- polarized hMDMs were incubated with 5 μg/mL PY265 or hIgG1 isotype for 3 hrs prior to inflammasome priming with 1 μg/mL LPS for 2.5 hrs. hMDMs were then stimulated with 1 mM ATP for 30 mins to activate inflammasome. Secreted IL-1β (pg/mL) was measured in conditioned media by MSD. Data are presented as mean values of technical triplicates ± (SD). *P*<0.05, using Tukey's multiple comparison test.

Anti-mouse MARCO Antibody, PY265m, Exhibits Anti-Tumor Activity as Single Agent and in Combination with Anti-PD1

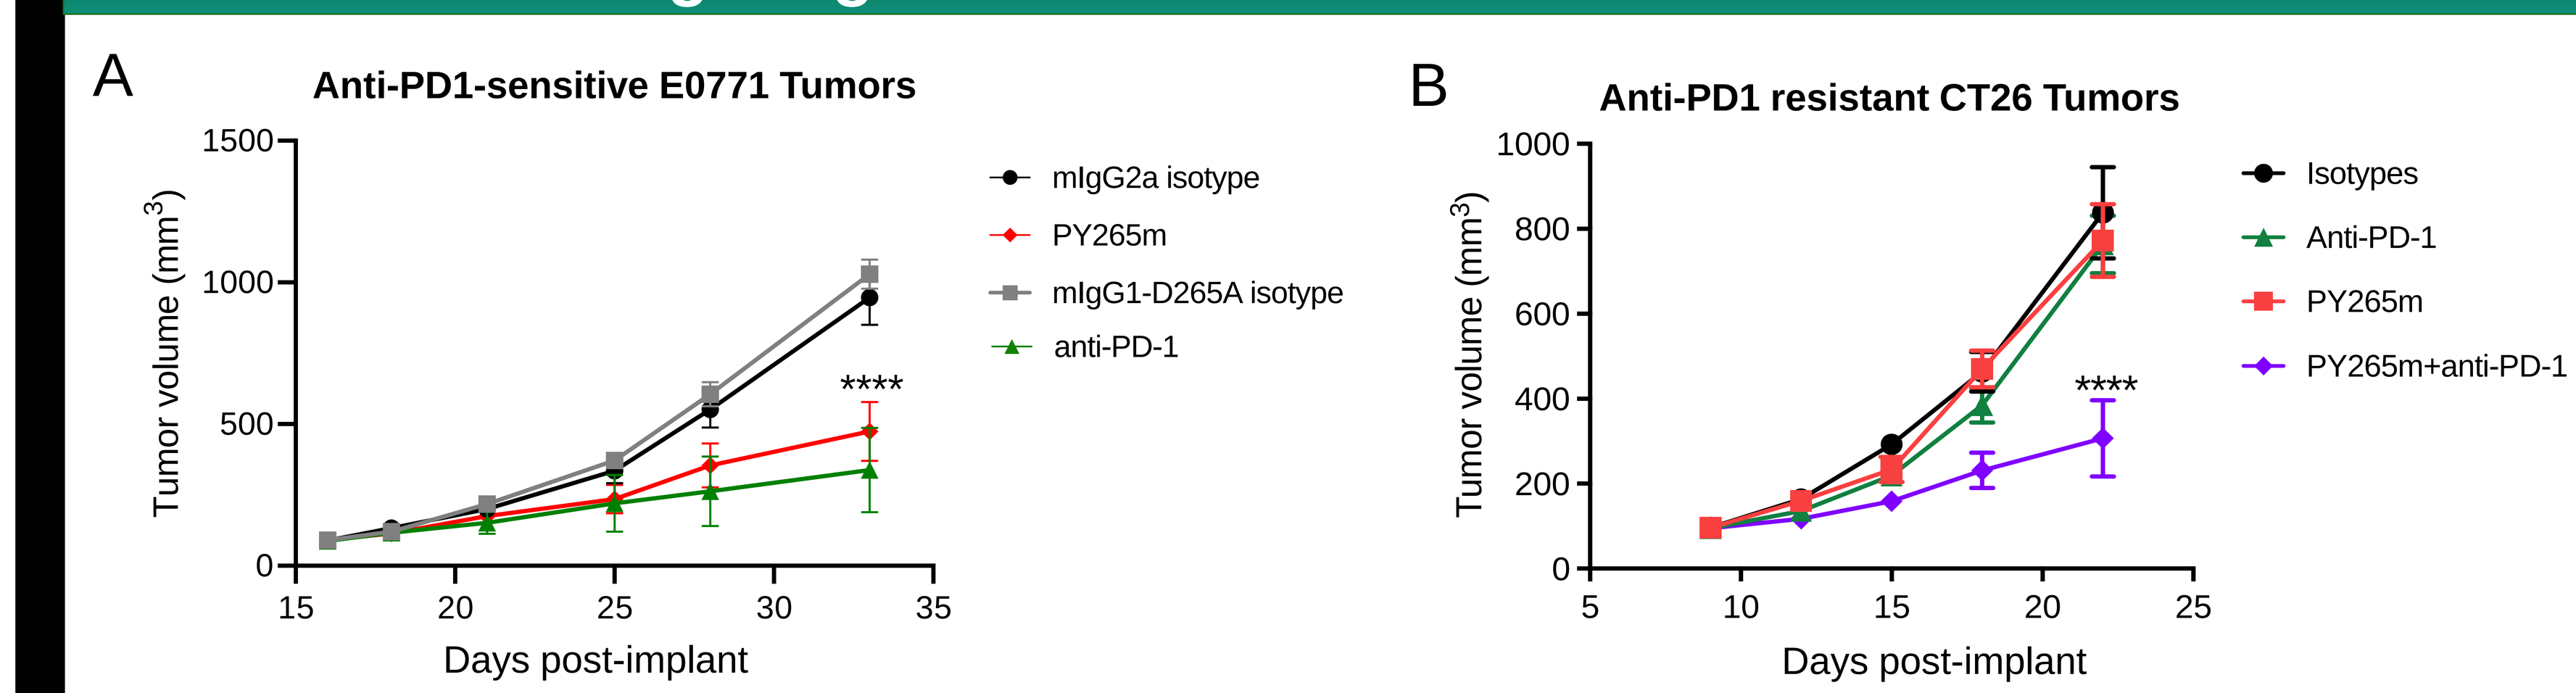
A
Anti-PD1-sensitive E0771 Tumors
Tumor volume (mm³)
Days post-implant
mIgG2a isotype
PY265m
mIgG1-D265A isotype
anti-PD-1

B
Anti-PD1 resistant CT26 Tumors
Tumor volume (mm³)
Days post-implant
Isotypes
Anti-PD-1
PY265m
PY265m+anti-PD-1

Figure 6. (A) Mice bearing established orthotopic E0771 syngeneic breast tumors were dosed with intraperitoneal (IP) injections of PY265m (10 mg/kg) and/or anti-PD-1 (5 mg/kg), or corresponding isotype control antibodies. Tumor volumes were measured at time points on the X axis, through Day 33. Anti-tumor efficacy of PY265m was significant when compared to its isotype (2-way ANOVA followed by Tukey's multiple comparisons test, *****P*<0.0001). **(B)** Mice bearing established CT26 syngeneic colorectal tumors were dosed with IP injections of anti-PD-1 (5 mg/kg) and/or PY265m (10 mg/kg), or isotype control antibodies. Tumor volumes were measured at timepoints on the X axis, through Day 22. The combination of PY265m and anti-PD-1 significantly reduced tumor growth vs anti-PD-1 alone, based on 2-way ANOVA followed by Tukey's multiple comparisons test (*****P*<0.0001).

Anti-MARCO Induces Proinflammatory Anti-Tumor Pathways In Vivo and Ex Vivo

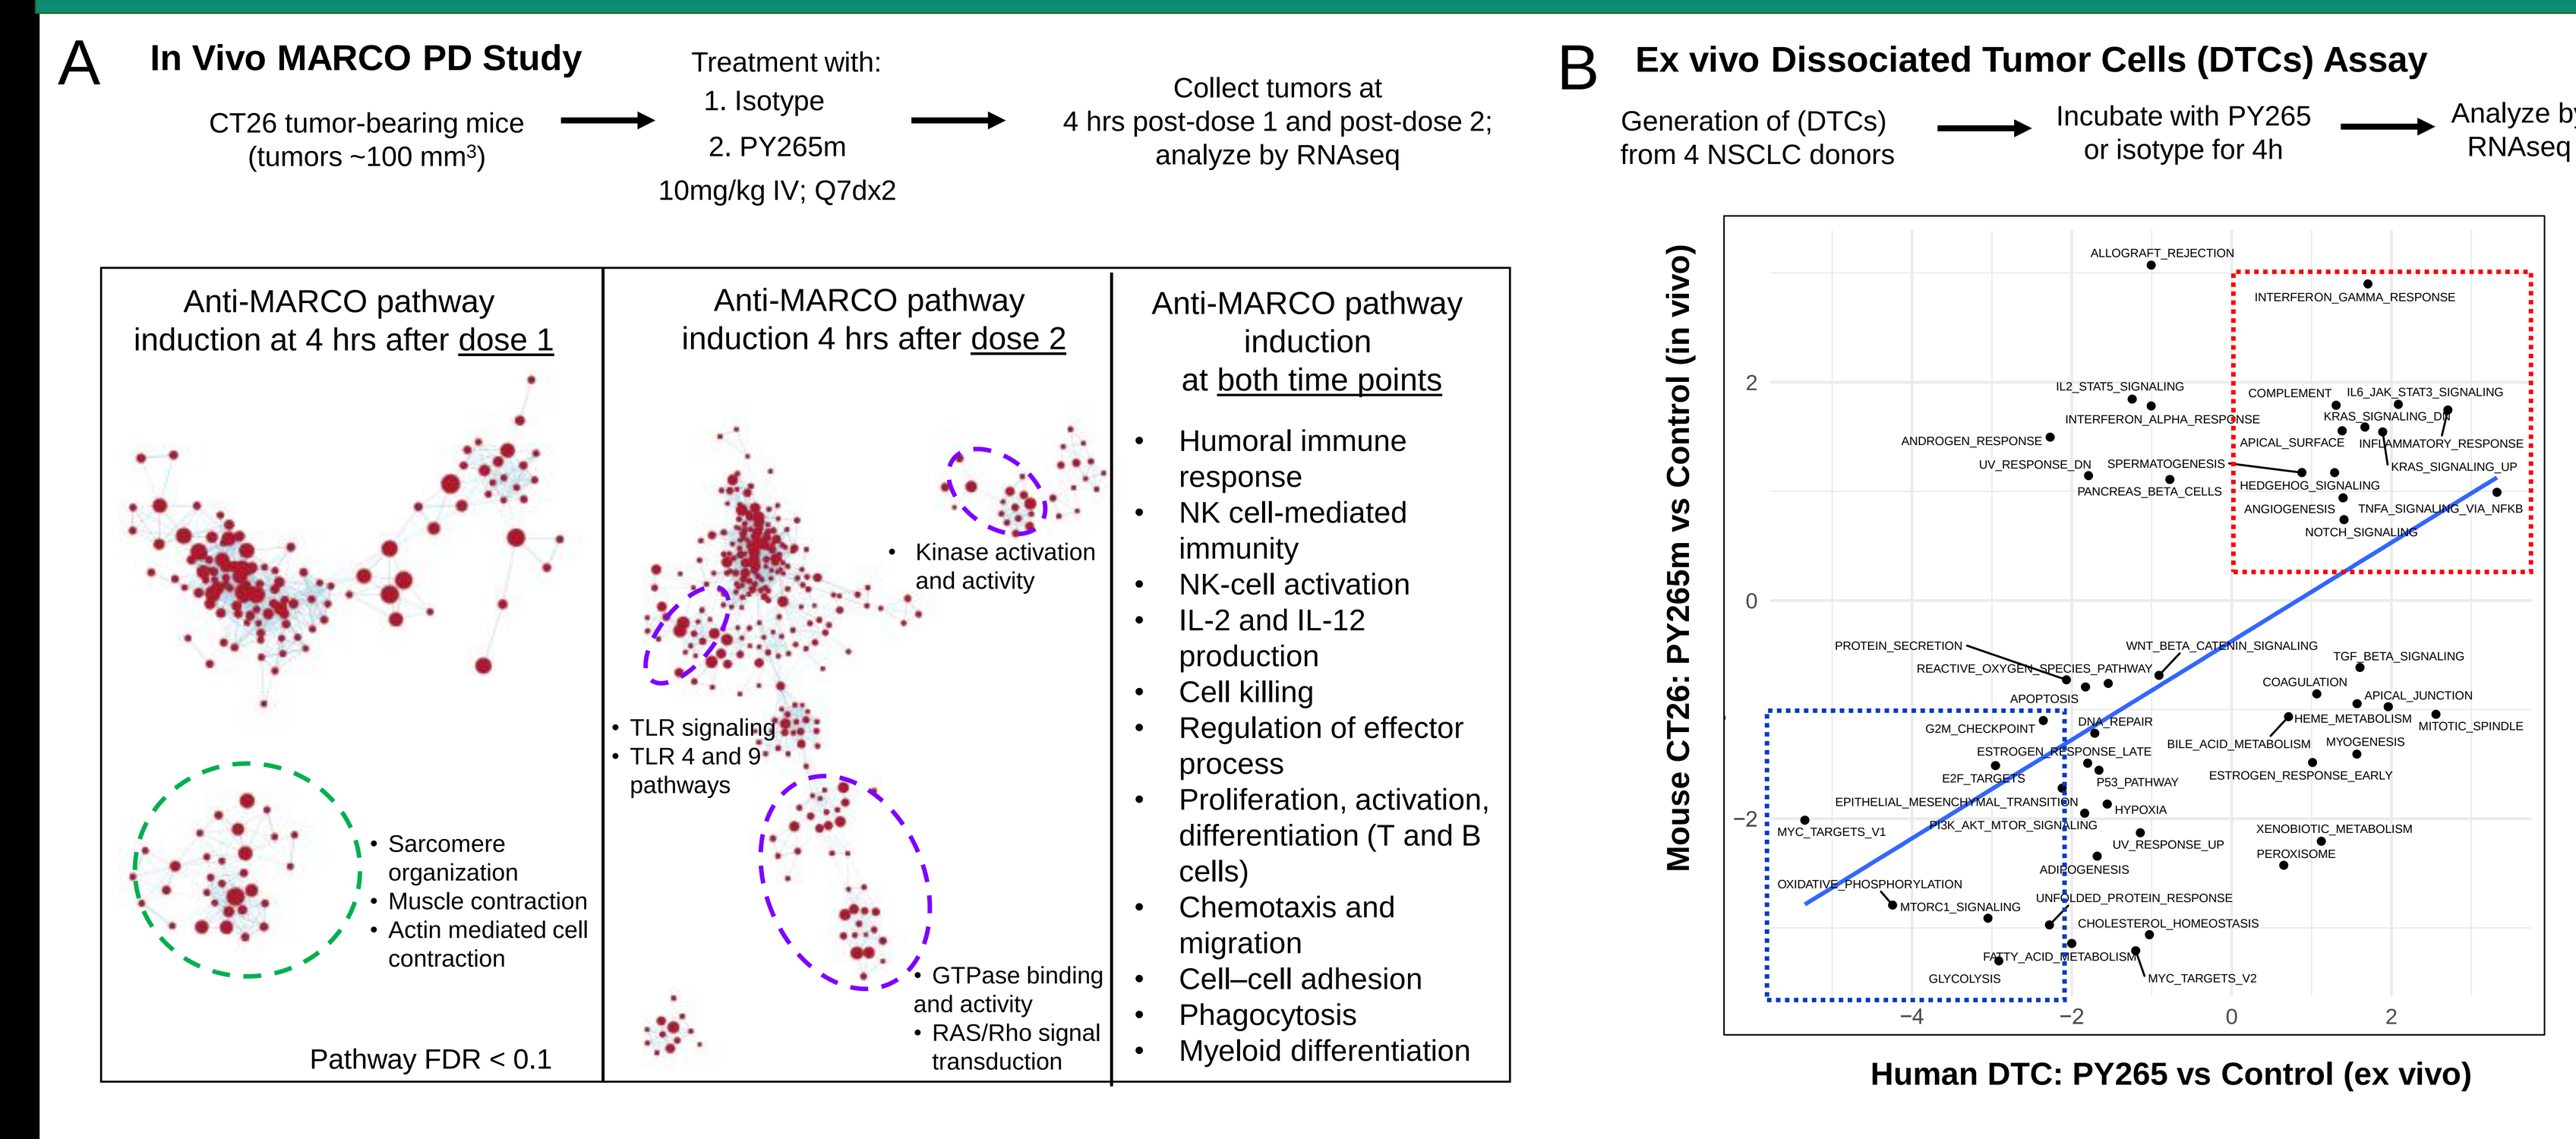
A
In Vivo MARCO PD Study
CT26 tumor-bearing mice (tumors ~100 mm³)
Treatment with:
1. Isotype
2. PY265m
10mg/kg IV; Q7dx2
Collect tumors at 4 hrs post-dose 1 and post-dose 2; analyze by RNAseq
Anti-MARCO pathway induction at 4 hrs after dose 1
Sarcomere organization
Muscle contraction
Actin mediated cell contraction
Pathway FDR < 0.1
Anti-MARCO pathway induction 4 hrs after dose 2
Kinase activation and activity
TLR signaling
TLR 4 and 9 pathways
GTPase binding and activity
RAS/Rho signal transduction
Anti-MARCO pathway induction at both time points
Humoral immune response
NK cell-mediated immunity
NK-cell activation
IL-2 and IL-12 production
Cell killing
Regulation of effector process
Proliferation, activation, differentiation (T and B cells)
Chemotaxis and migration
Cell–cell adhesion
Phagocytosis
Myeloid differentiation
B
Ex vivo Dissociated Tumor Cells (DTCs) Assay
Generation of (DTCs) from 4 NSCLC donors
Incubate with PY265 or isotype for 4h
Analyze by RNAseq
Mouse CT26: PY265m vs Control (in vivo)
Human DTC: PY265 vs Control (ex vivo)

Figure 7. (A) RNA was extracted from CT26 tumors from mice given injections of PY265m or isotype control mAb (single-dose PD study) and sequenced. Differentially regulated pathways are highlighted as spider plots for significant pathways at Dose 1 only (green circle), Dose 2 only (purple circles), and both time points. **(B)** Total RNA was isolated from non-small cell lung cancer (NSCLC) DTCs incubated with PY265 or hIgG1 (isotype control) and sequenced. GSEA-derived enrichment scores from mouse (in vivo) DTC (ex vivo) experiments indicated that the human PY265 mAb and mouse surrogate PY265m induced common proinflammatory and immunostimulatory pathways (red square) and decreased common survival and tumor-growth associated pathways (blue square).

PY265m Elicits Anti-Tumor Activity and Reprogramming of TAMs and mMDSCs with Rapid Activation of Intra-tumoral Immunity

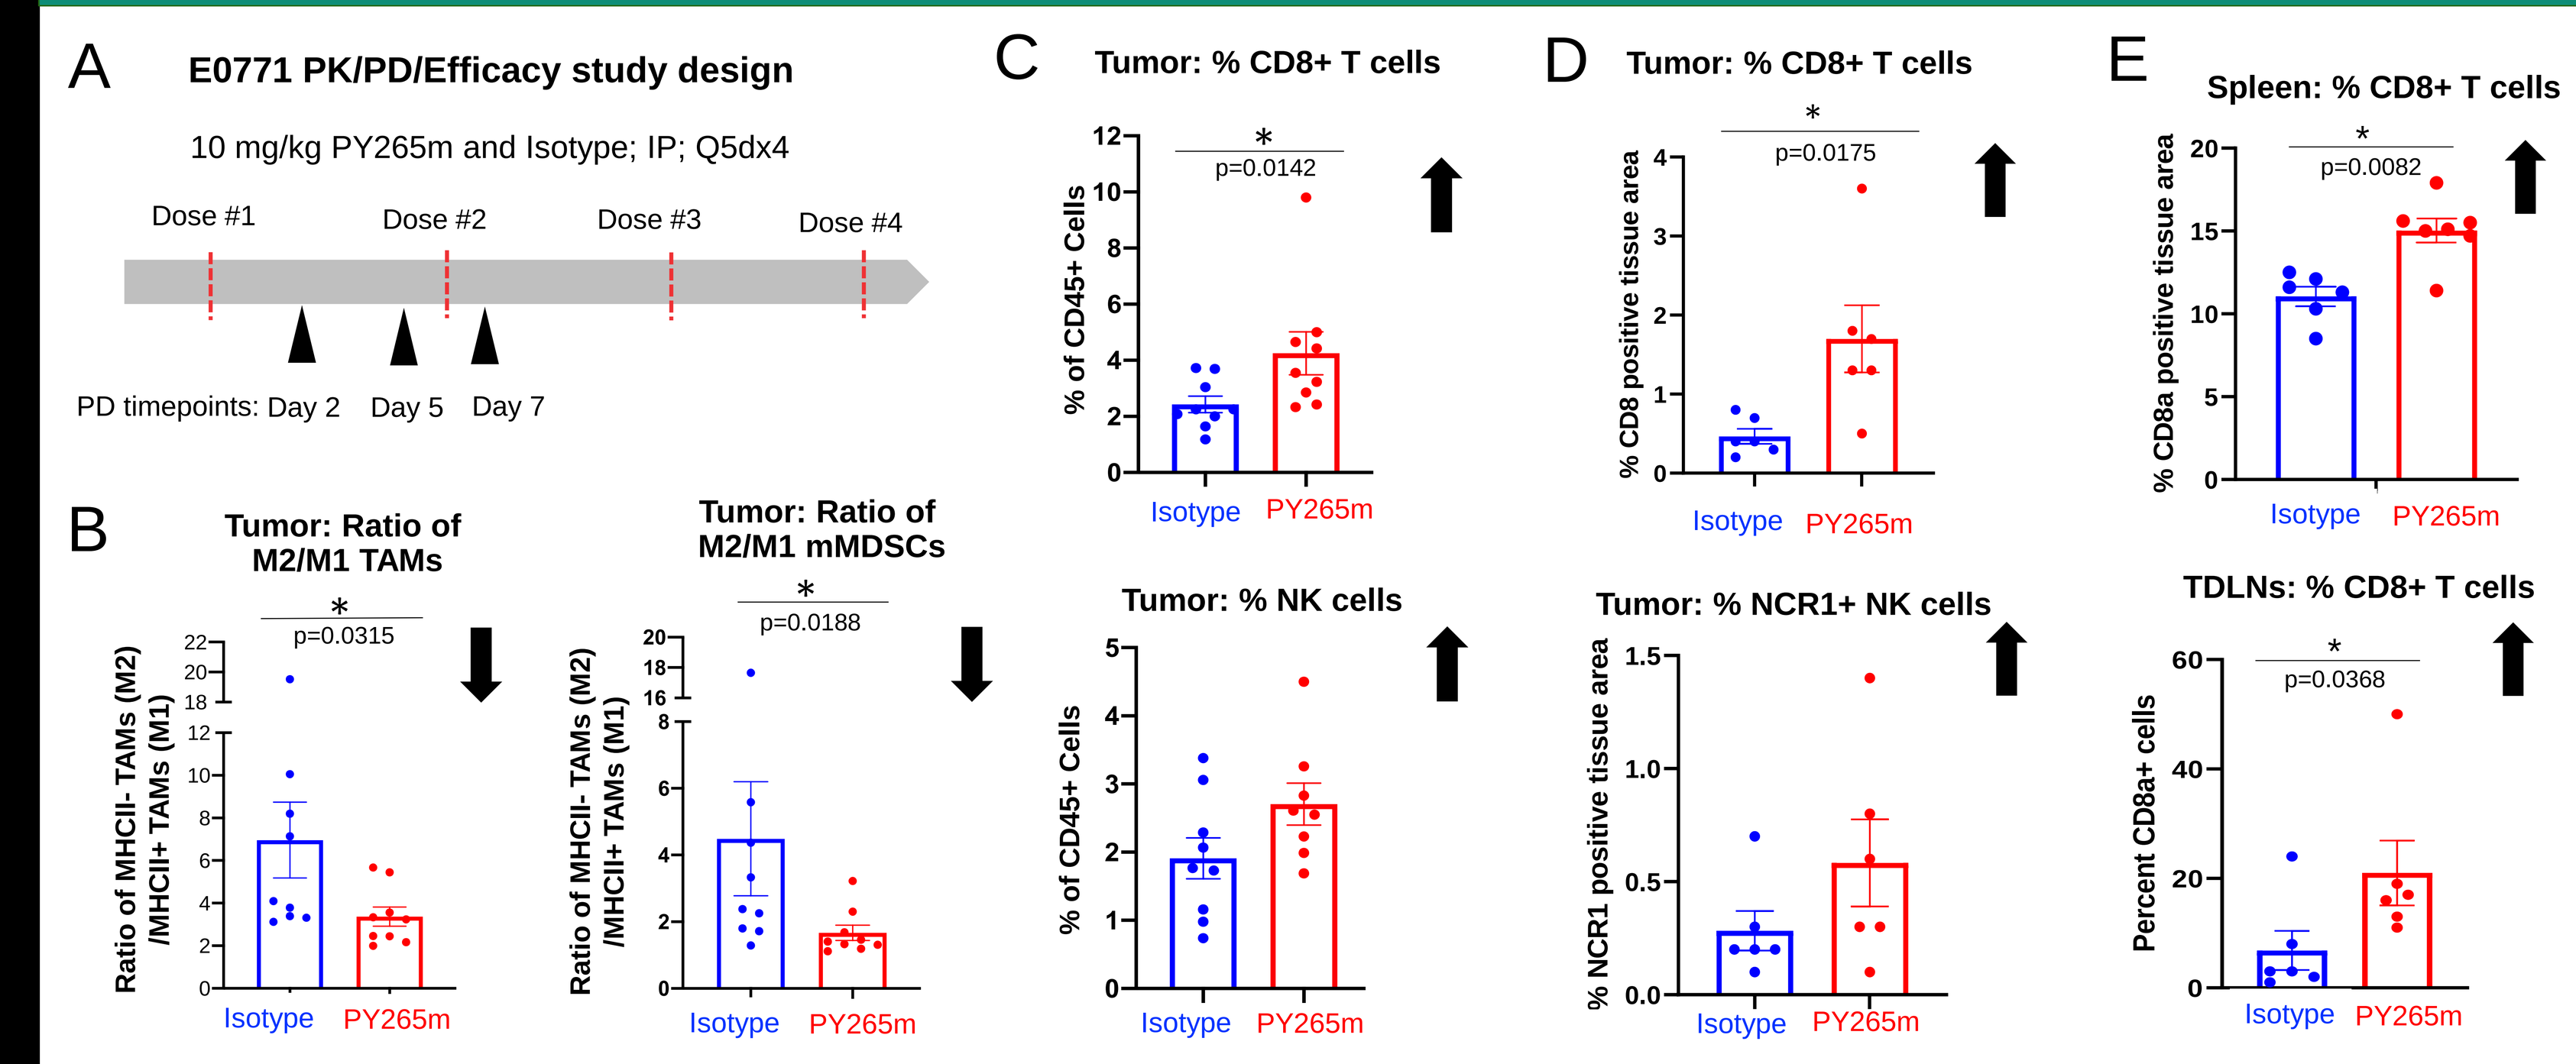
A
E0771 PK/PD/Efficacy study design
10 mg/kg PY265m and Isotype; IP; Q5dx4
Dose #1
Dose #2
Dose #3
Dose #4
PD timepoints: Day 2 Day 5 Day 7
B
Tumor: Ratio of M2/M1 TAMs
p=0.0315
Tumor: Ratio of M2/M1 mMDSCs
p=0.0188
C
Tumor: % CD8+ T cells
p=0.0142
Tumor: % NK cells
D
Tumor: % CD8+ T cells
p=0.0175
Tumor: % NCR1+ NK cells
E
Spleen: % CD8+ T cells
p=0.0082
TDLNs: % CD8+ T cells
p=0.0368
Isotype
PY265m

Figure 7. (**A**) PY265m PK, PD, and efficacy study in mice with E0771 orthoptic tumors given IP injections of PY265m or isotype control mAb; samples were collected at 3 timepoints for PD analysis from a subset of mice to assess myeloid and lymphoid changes. (**B**) Flow cytometry data showing differences in myeloid populations (as M1/M2 ratios on Day 2), and **(C)** CD8+ cells (Day 2), and NK cells (Day 5), as a frequency of CD45+ cells. **(D)** IHC data from tumor tissues collected at Day 7 after dose 2. Image analysis was used to quantify the percentages of CD8+ T cells (top) and NCR1+ cells (bottom) over the whole tissue area. **(E)** IHC image analysis data showing the percentage CD8+ T cells (Day 5) in the marginal zone of the spleen (top) and in the paracortical regions of tumor-draining lymph nodes (TDLNs) (bottom). Data are presented as mean percentage values from 6 mice within each group ± standard error of the mean (SEM). Mann Whitney unpaired *t* test was used for statistical analysis.

Proposed Mechanism for PY265 Activity

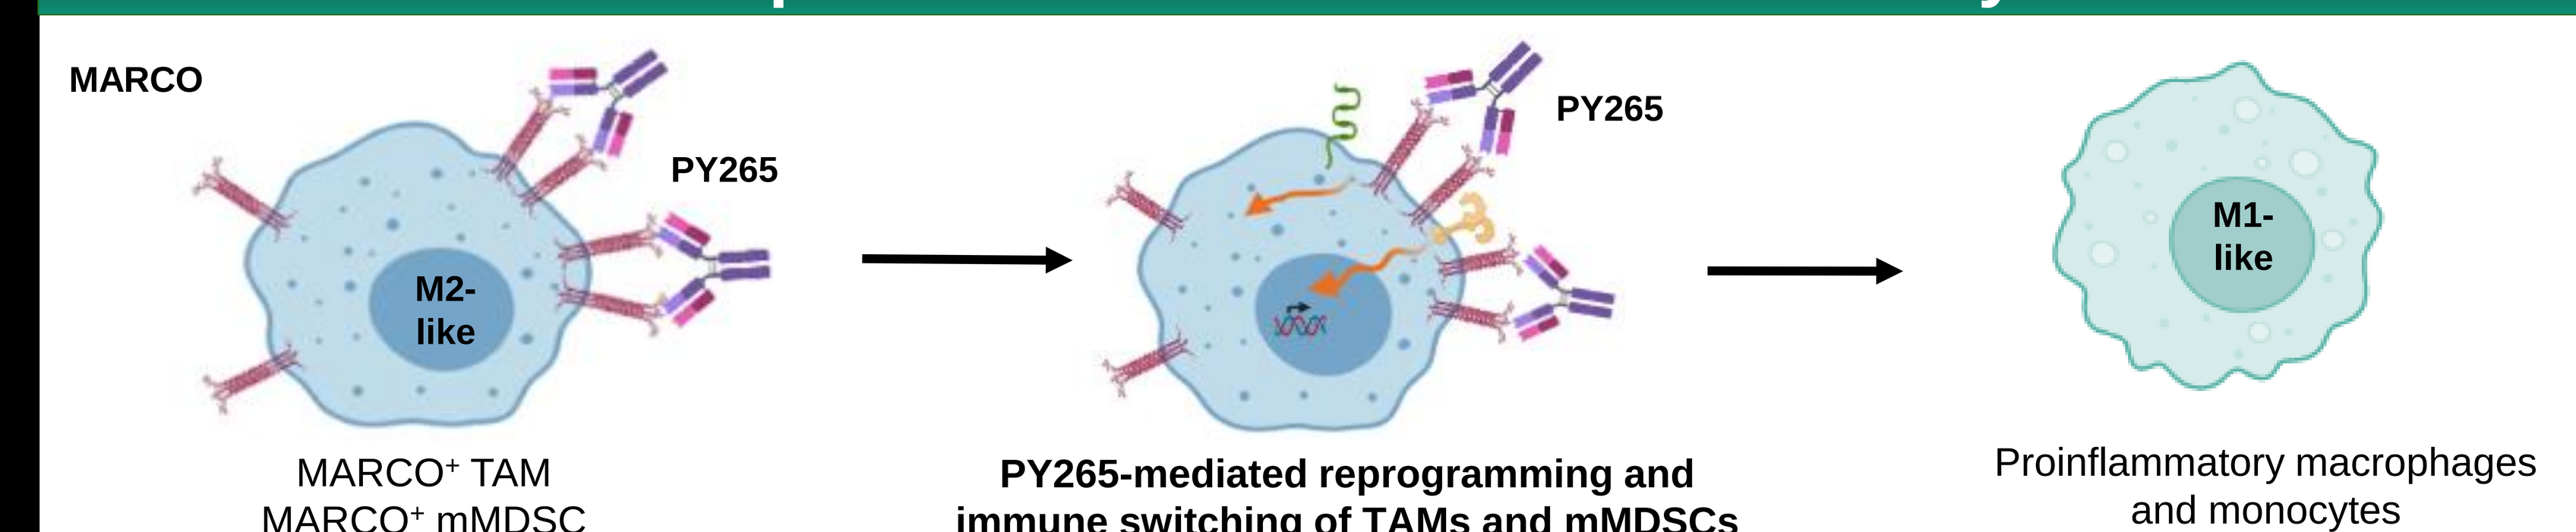
MARCO
PY265
M2-like
PY265
M1-like

MARCO⁺ TAM
MARCO⁺ mMDSC

Immunosuppression
- Upregulated in IL-10 enriched and hypoxic TME
- Suppresses immune cytotoxic activity
- Increases angiogenesis and metastasis

PY265-mediated reprogramming and immune switching of TAMs and mMDSCs
- Rapid modulation of phospho-signaling cascades (RTK, STAT1, inflammatory and NF-κB signaling, cell cycle & apoptosis, cell adhesion and cytoskeletal rearrangement)
- Changes in phagocytosis, cell adhesion, motility, and transcription
- Activation of NF-κB reporter activity as single agent and in combination with TLRs agonists
- Induction of pro-inflammatory cytokines and chemokines secretion
- Increase inflammasome activation

Proinflammatory macrophages and monocytes

Anti-tumor immunity
- Increases and activates CD8+ T cells and NK cells
- Recruits proinflammatory myeloid cells
- Increases antigen presentation

- PY265 is safe and generally well tolerated in pilot NHP toxicology studies.
- Terminal half-life of PY265 PK is typical of mAbs.
- No significant cytokine release observed in whole blood or PBMCs in response to PY265.
- PY265 first in human clinical trials are planned for 2023 in patients with advanced solid tumors refractory or relapsed to standard care, as single agent and in combination with CPI.